Macrophage migration inhibitory factor (MIF) involvement in breast cancer cell metabolism: A ¹H-NMR spectroscopy evaluation

V. Richard[1-4]*, R. Conotte[2,4], N. Kindt[3], S. Saussez[3], J-M. Colet[2,4]

[1] CHU Ambroise Paré, Mons Belgium ; [2] Department of Human Biology and Toxicology, Faculty of Medicine and Pharmacy, UMONS, Mons, Belgium;
[3] Department of Anatomy and Cell Biology, Faculty of Medicine and Pharmacy, UMONS, Mons, Belgium ; [4] Biomedical Profiling Unit, UMHAP, Mons, Belgium
*e-mail: vincent.richard@hap.be

Background

Breast cancer (BC) is the most common diagnosed cancer and the leading cause of cancer death in women worldwide, accounting for 25% of all cancer cases and 15% of all cancer deaths among females. There is an obvious need for a better understanding of BC biology.

The shift in cellular metabolism, also called Warburg effect, is an emerging hallmark of cancer cells (CC) orchestrated by oncogenic proteins. MIF, a pleiotropic cytokine, is involved in BC cells proliferation and multiple aspects of carcinogenesis but little is known about its possible role in cellular metabolic activities.

Metabolomics, the global qualitative and quantitative evaluation of metabolites in a biological system, by NMR spectroscopy, mass spectrometry or combined techniques, has emerged as a unique tool to investigate the modification of metabolites of cancer cells or in bio fluids and tissues of cancer patients.

Using MIF knockdown (KD) technique and ¹H-RMN spectroscopy, we evaluated the influence of MIF on BC triple receptor negative MDA-MB-231 and hormones receptors positive MCF-7 metabolism cells lines in the hope of identifying therapeutic targets.

Methods

Metabonomic strategy

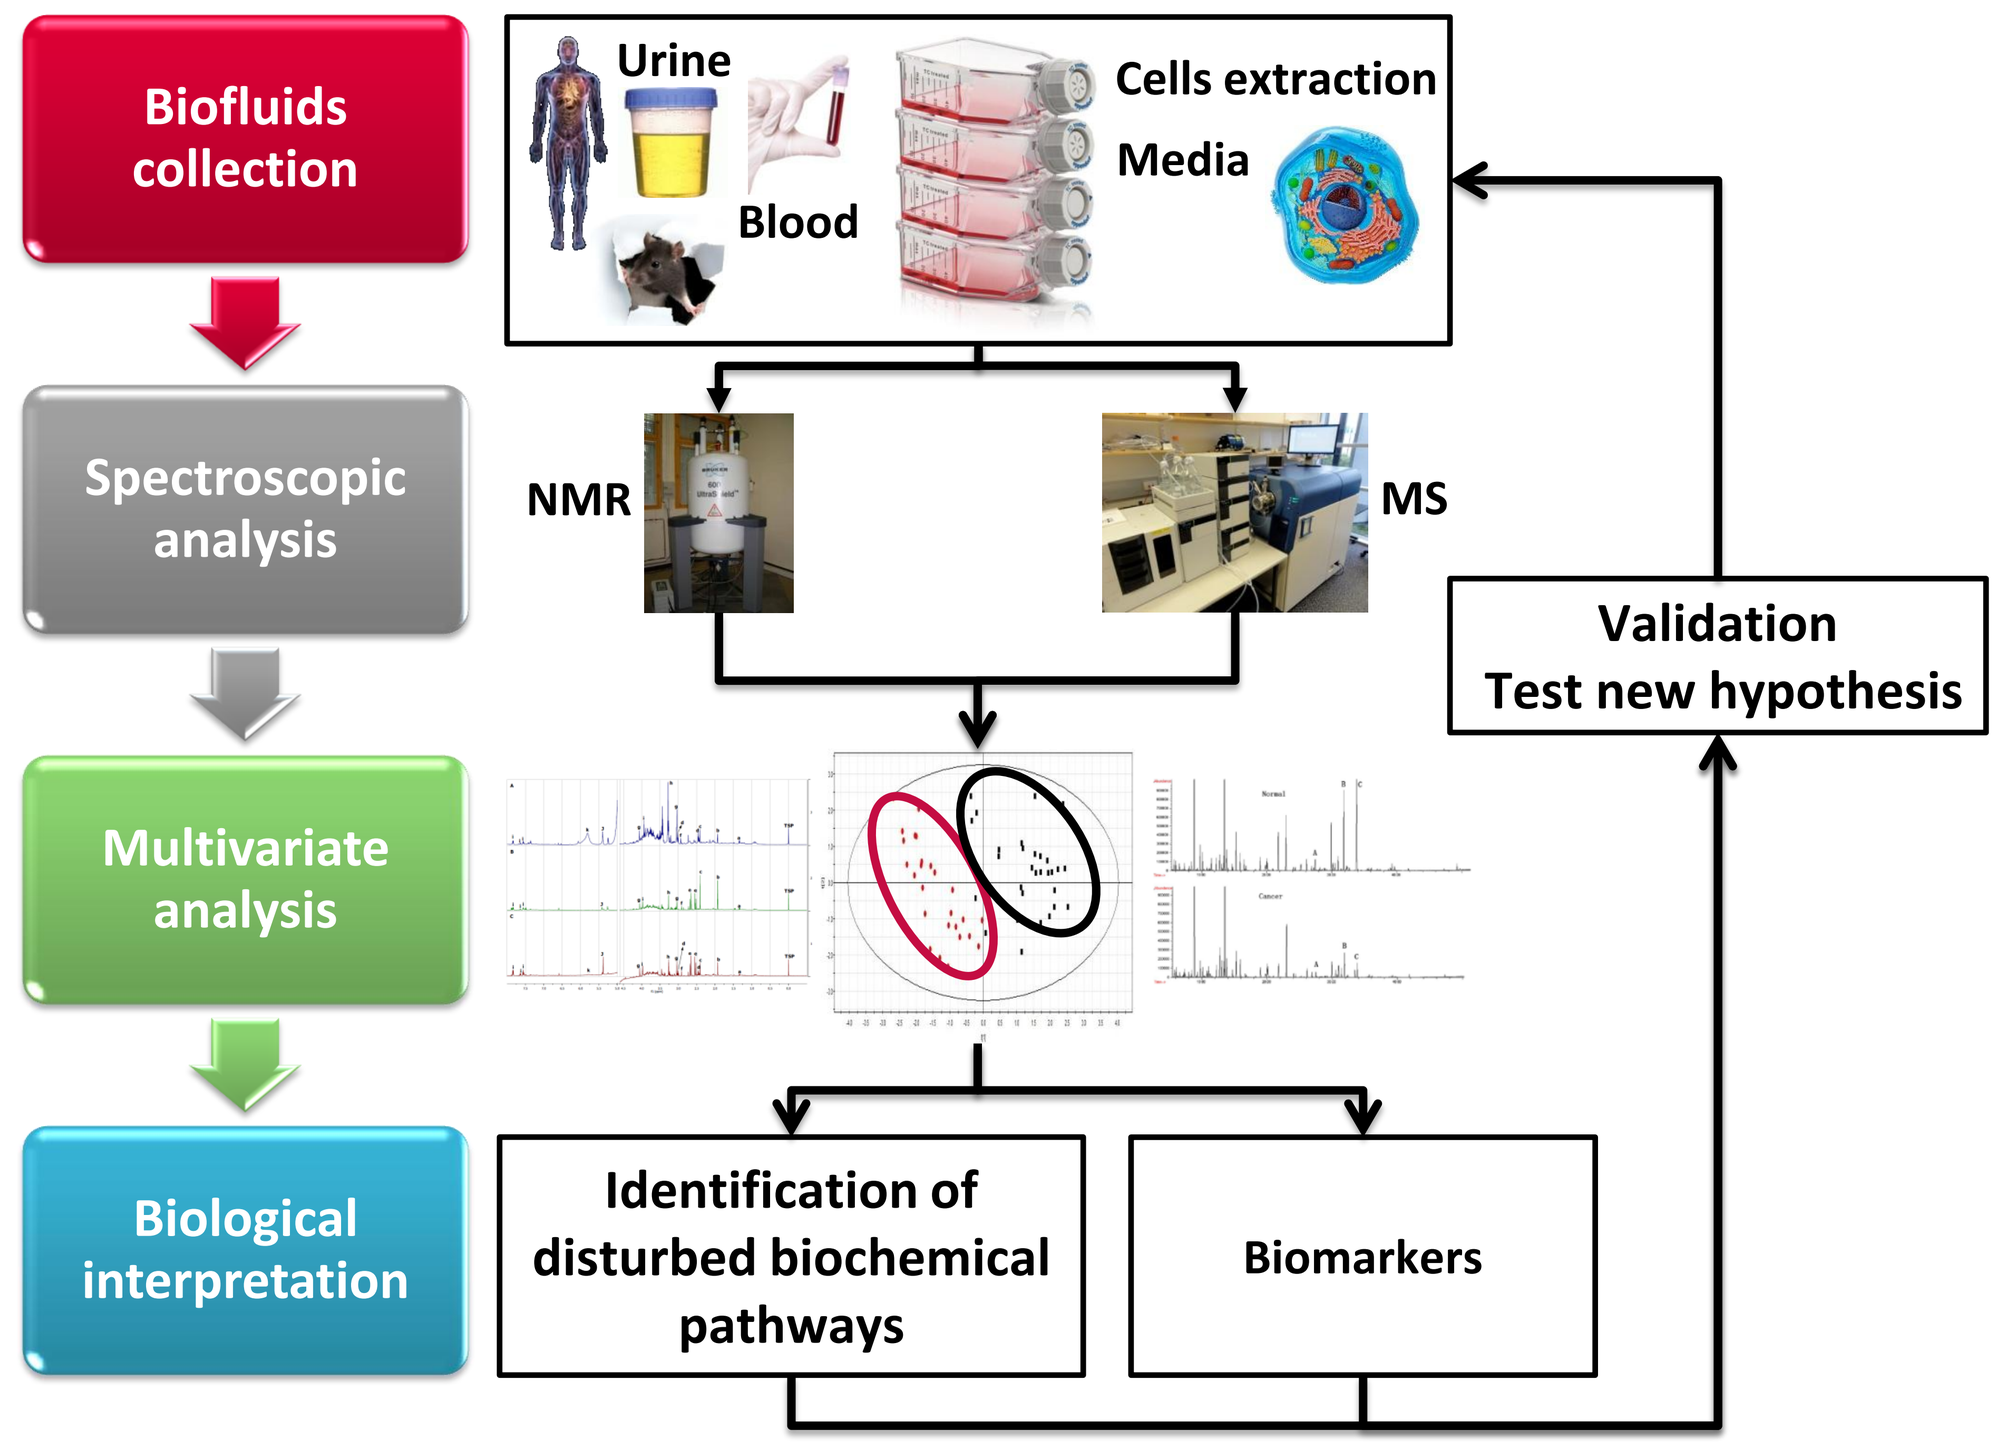
Biofluids collection
Urine
Blood
Cells extraction
Media
Spectroscopic analysis
NMR
MS
Validation
Test new hypothesis
Multivariate analysis
Biological interpretation
Identification of disturbed biochemical pathways
Biomarkers

Cell culture

MIF KD cells were obtained using MIF shRNA lentiviral particles.

The Western Blot below shows a decrease in MIF expression in MCF-7 and MDA-MB-231 knock-down (KD) cell lines compared to control (sc).

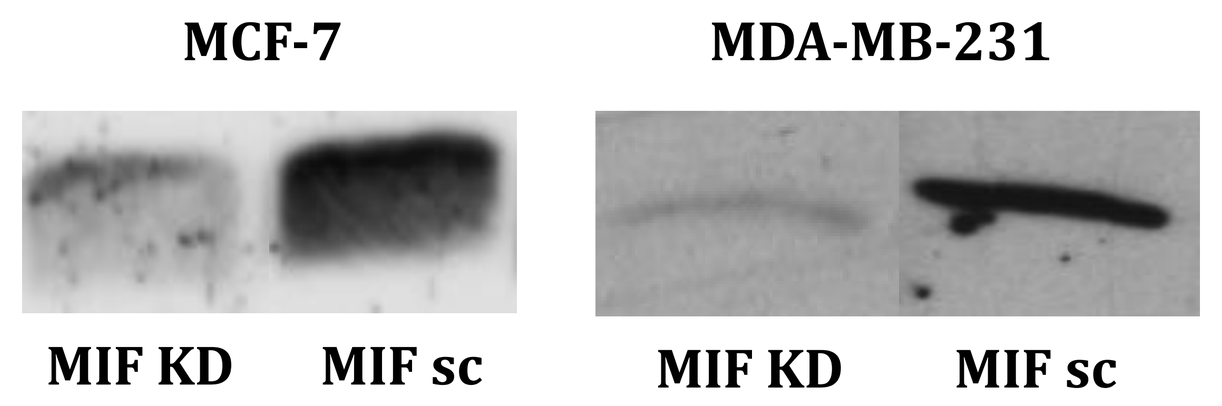
MCF-7
MDA-MB-231
MIF KD
MIF sc
MIF KD
MIF sc

Cells were cultured in DMEM with serum during 4 days and maintained in DMEM serum free during 48h before extraction.

Metabolites extraction

After a quick rinsing, cells were scrapped and quenched using cold methanol.
The solution containing the quenched cells was pipetted for intracellular metabolites extraction (methanol, chloroform and water extraction procedure).
In this study, only the aqueous phase containing water soluble low-molecular-weight endogenous metabolites was used. Prior to ¹H-NMR analysis, solvents were completely removed using a vacuum concentrator. Before acquisition on a Bruker Avance spectrometer at 500 MHz, each sample was reconstituted in phosphate buffer mixed with TSP.

Spectra handling & Multivariate analysis

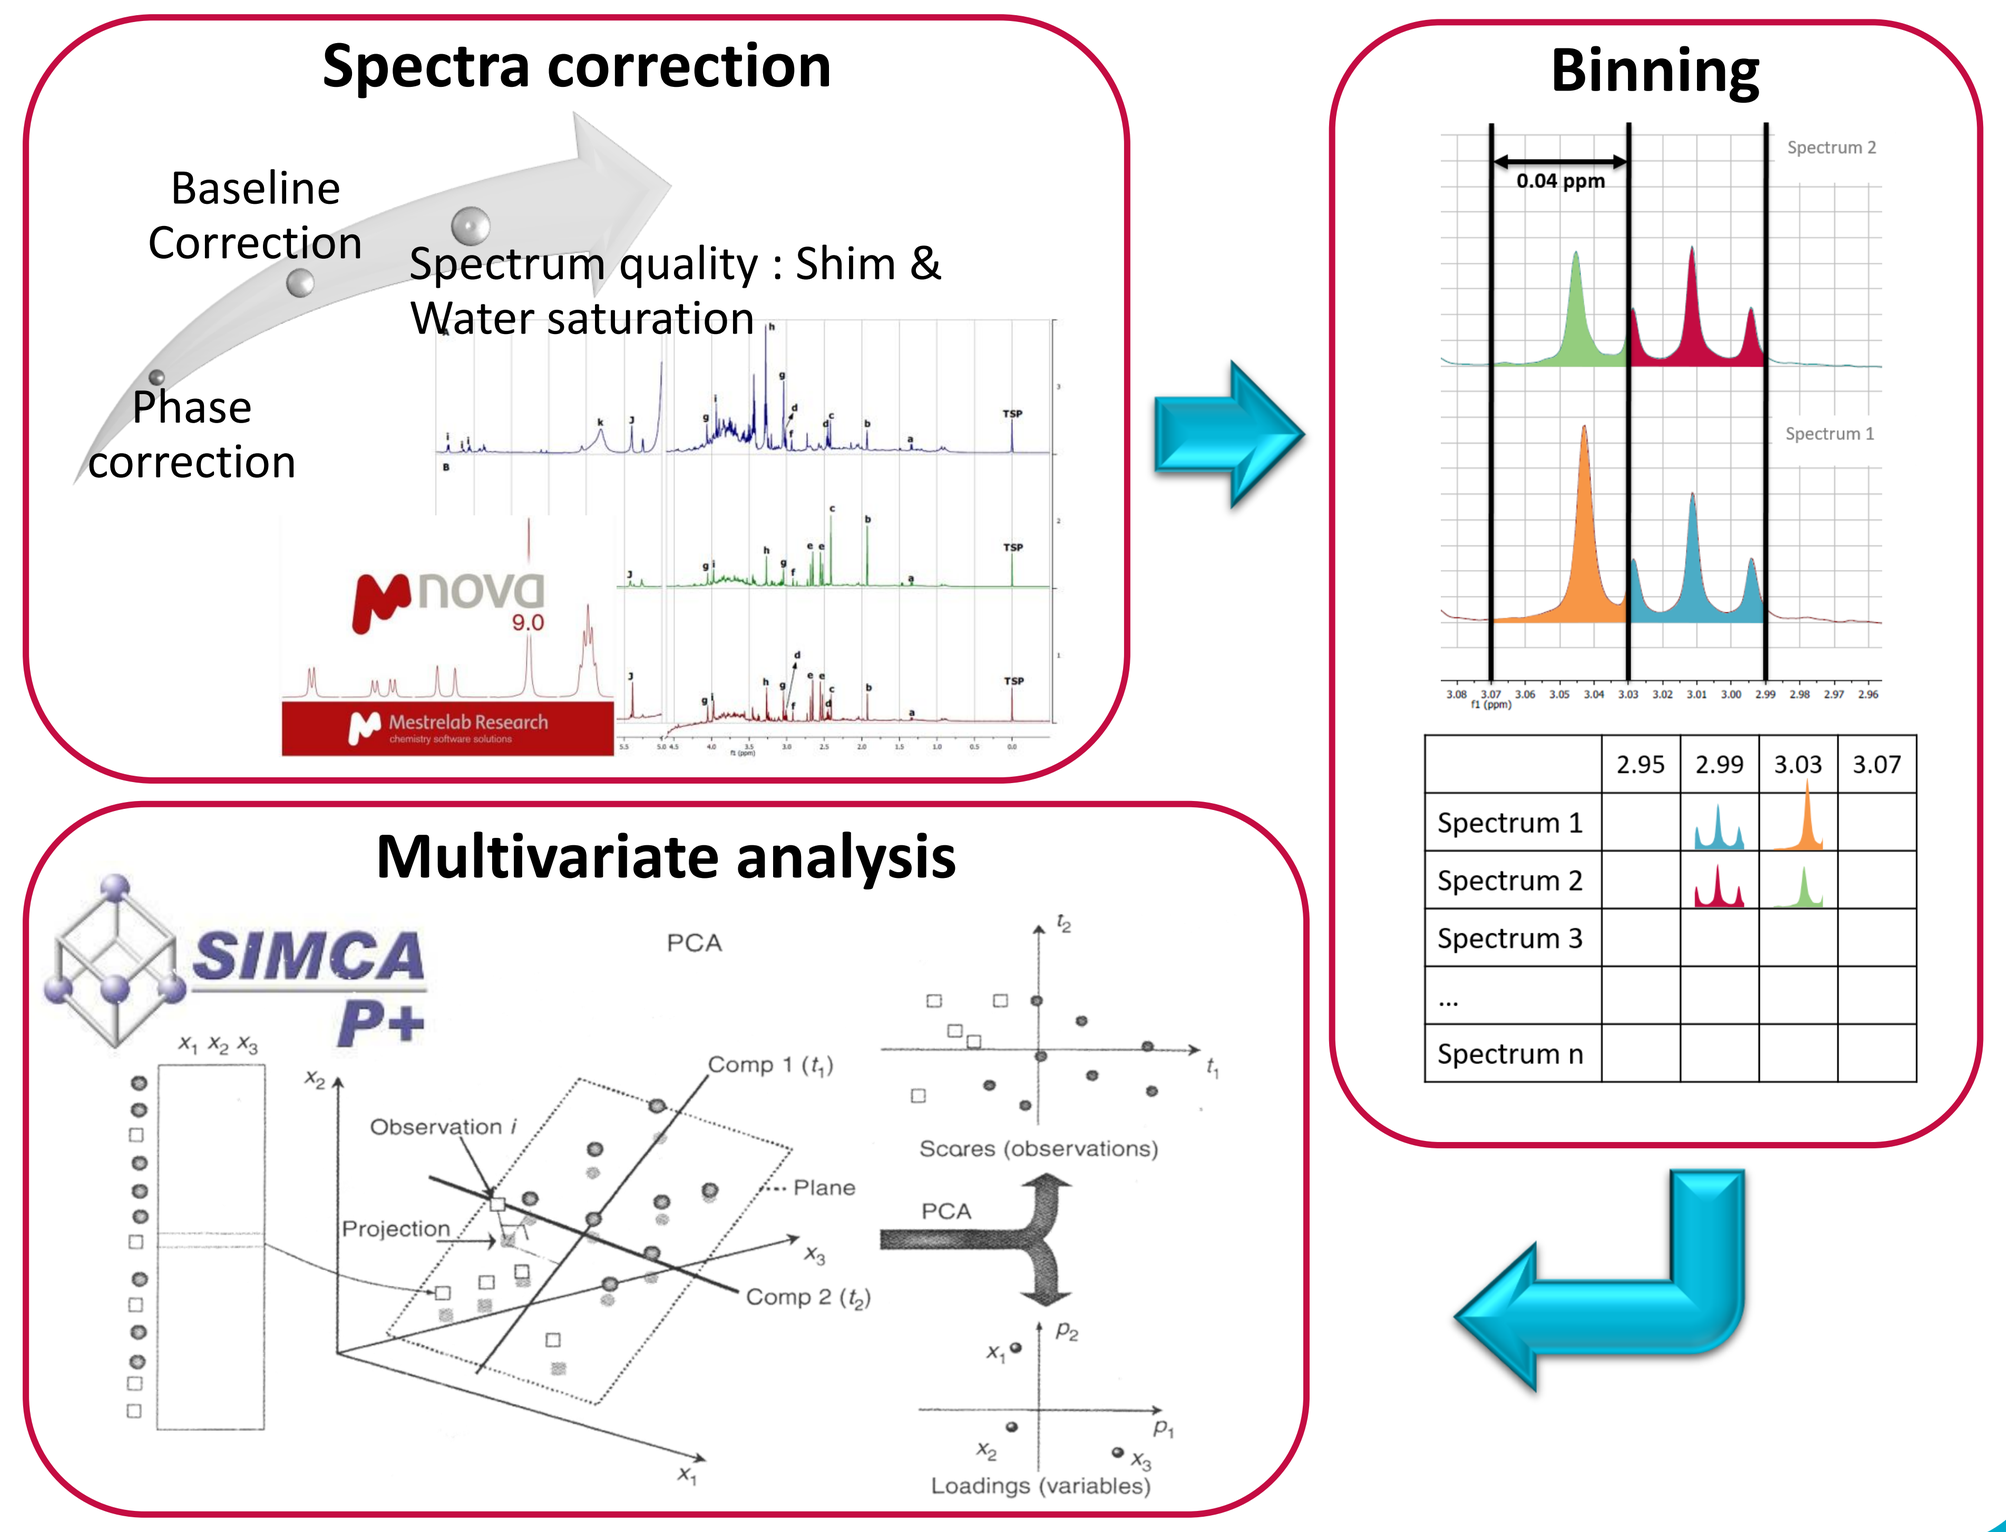
Spectra correction
Baseline Correction
Spectrum quality : Shim & Water saturation
Phase correction
Binning
0.04 ppm
Spectrum 2
Spectrum 1
2.95
2.99
3.03
3.07
Spectrum 1
Spectrum 2
Spectrum 3
...
Spectrum n
Multivariate analysis
SIMCA P+
PCA
Comp 1 (t1)
Comp 2 (t2)
Observation i
Projection
Plane
Scores (observations)
Loadings (variables)

Results

MCF7 vs MDA-MB-231

1A

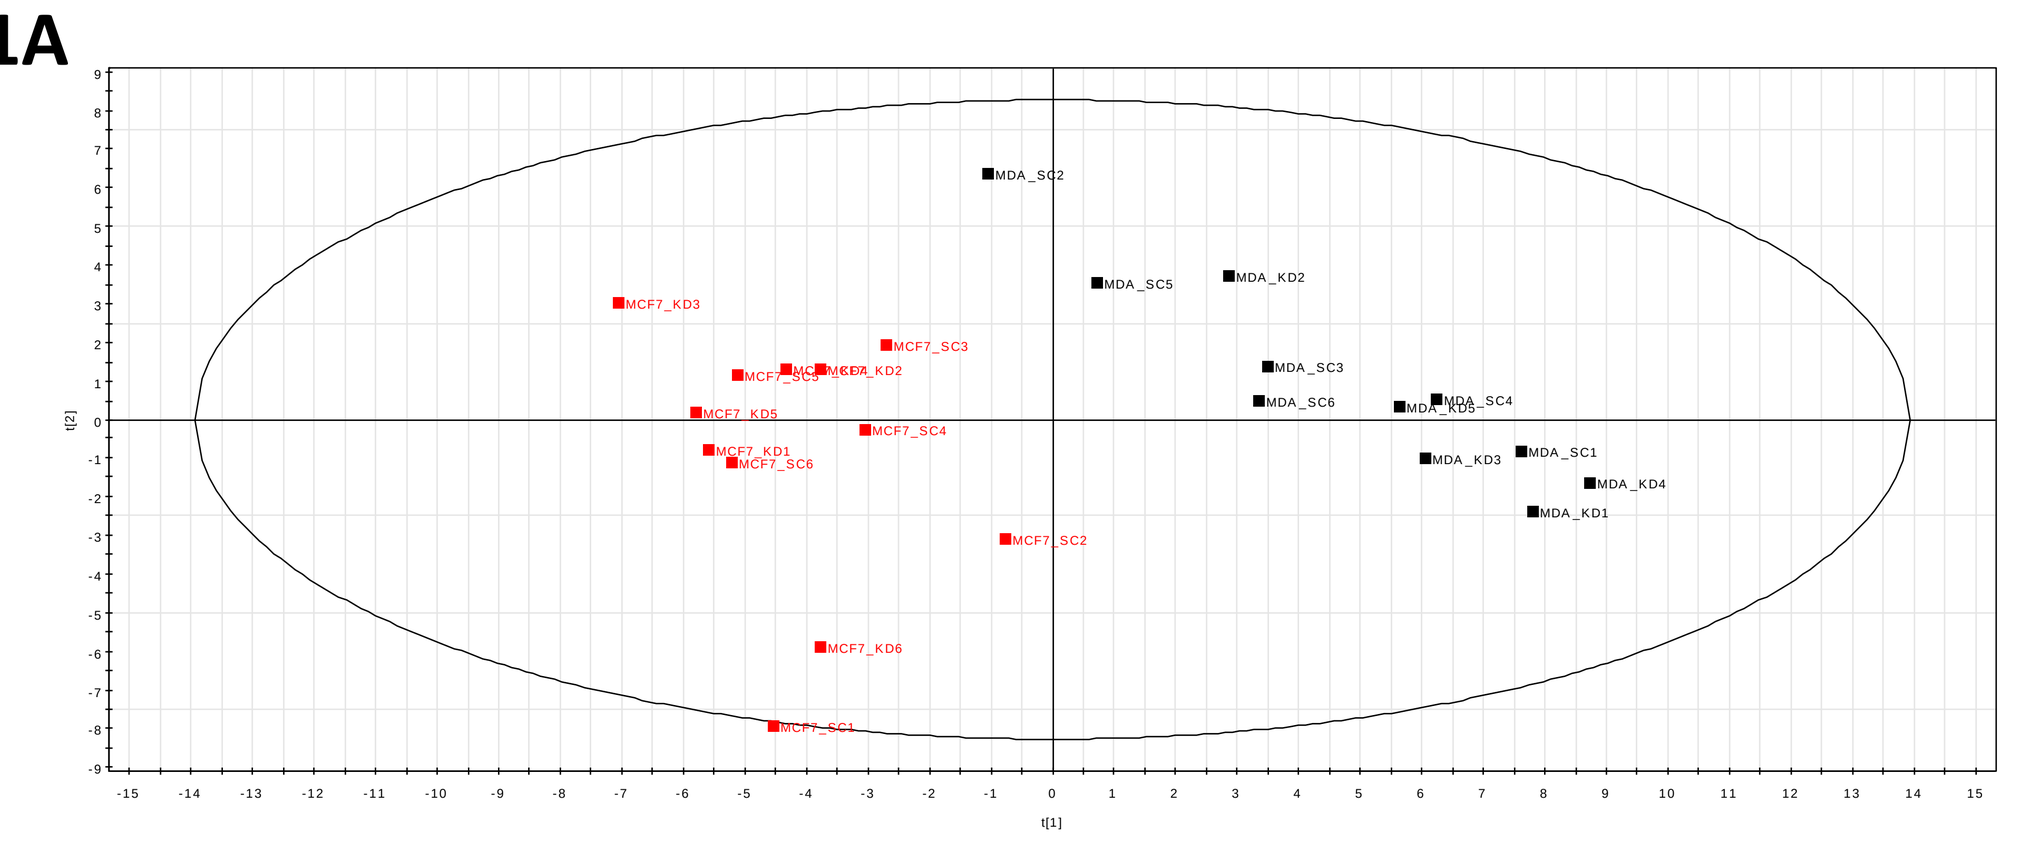

1B

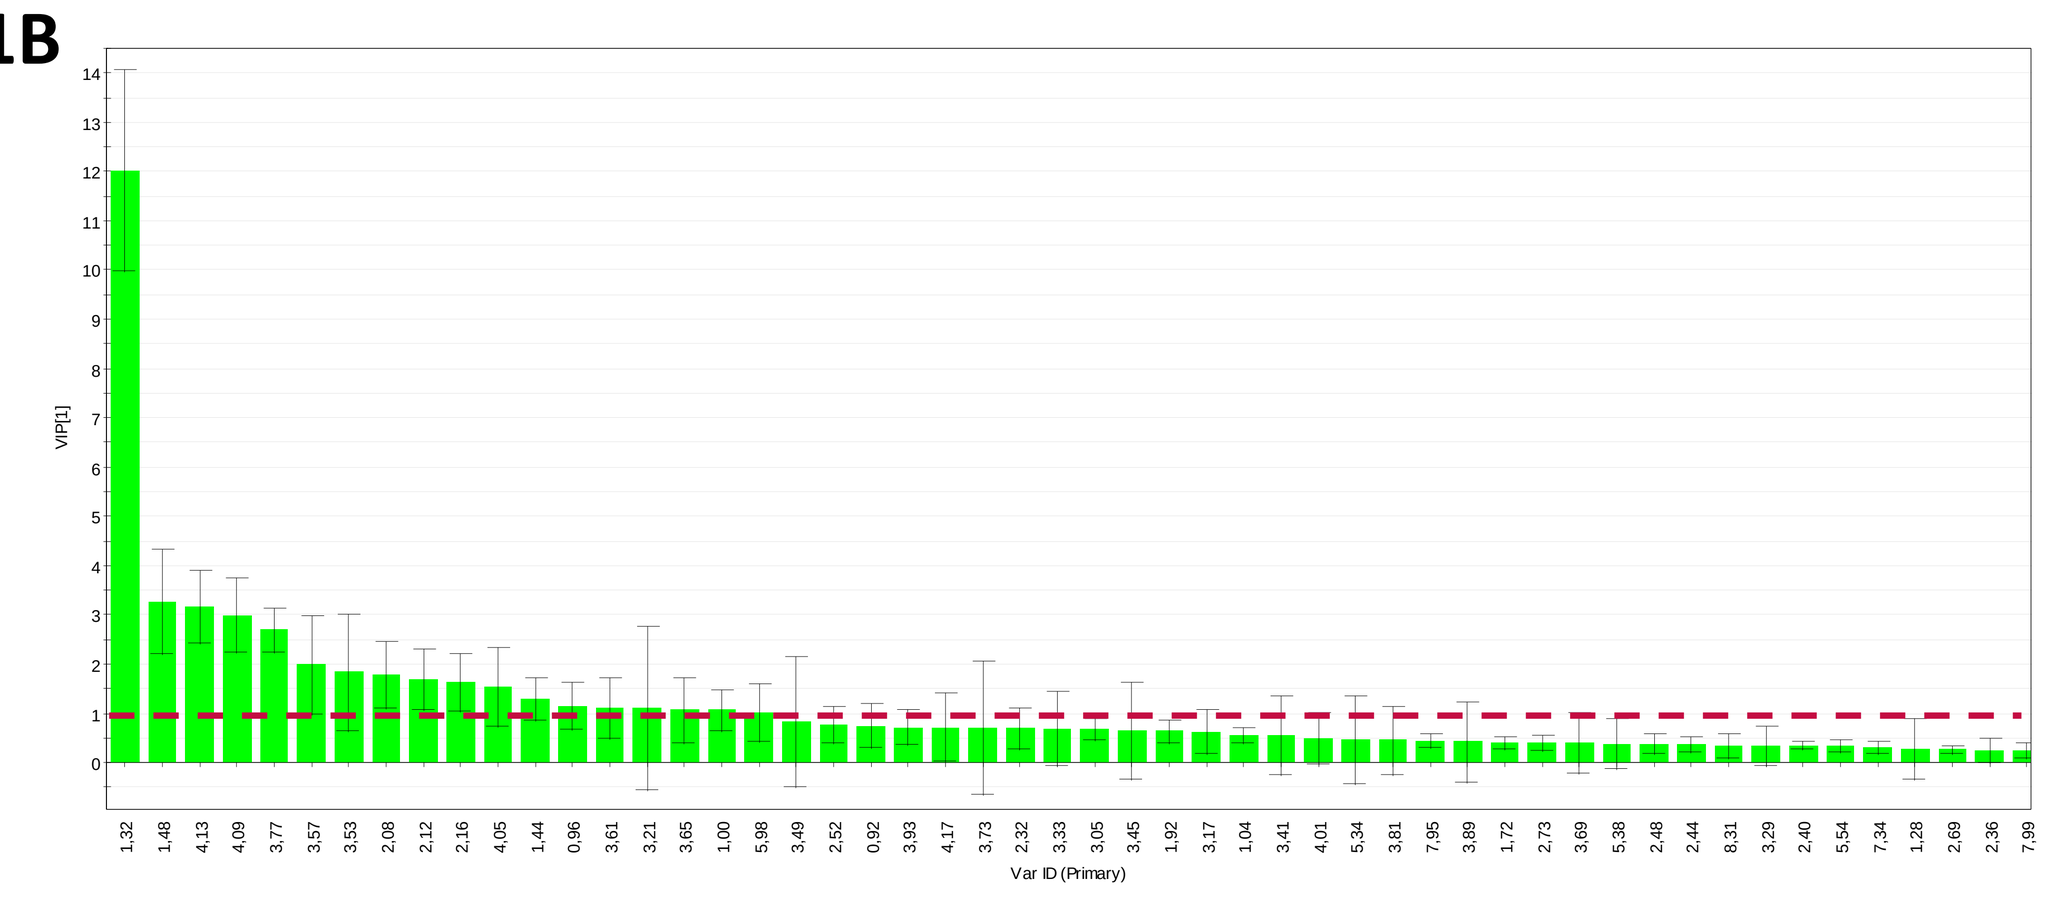
VIP[1]
Var ID (Primary)

1C

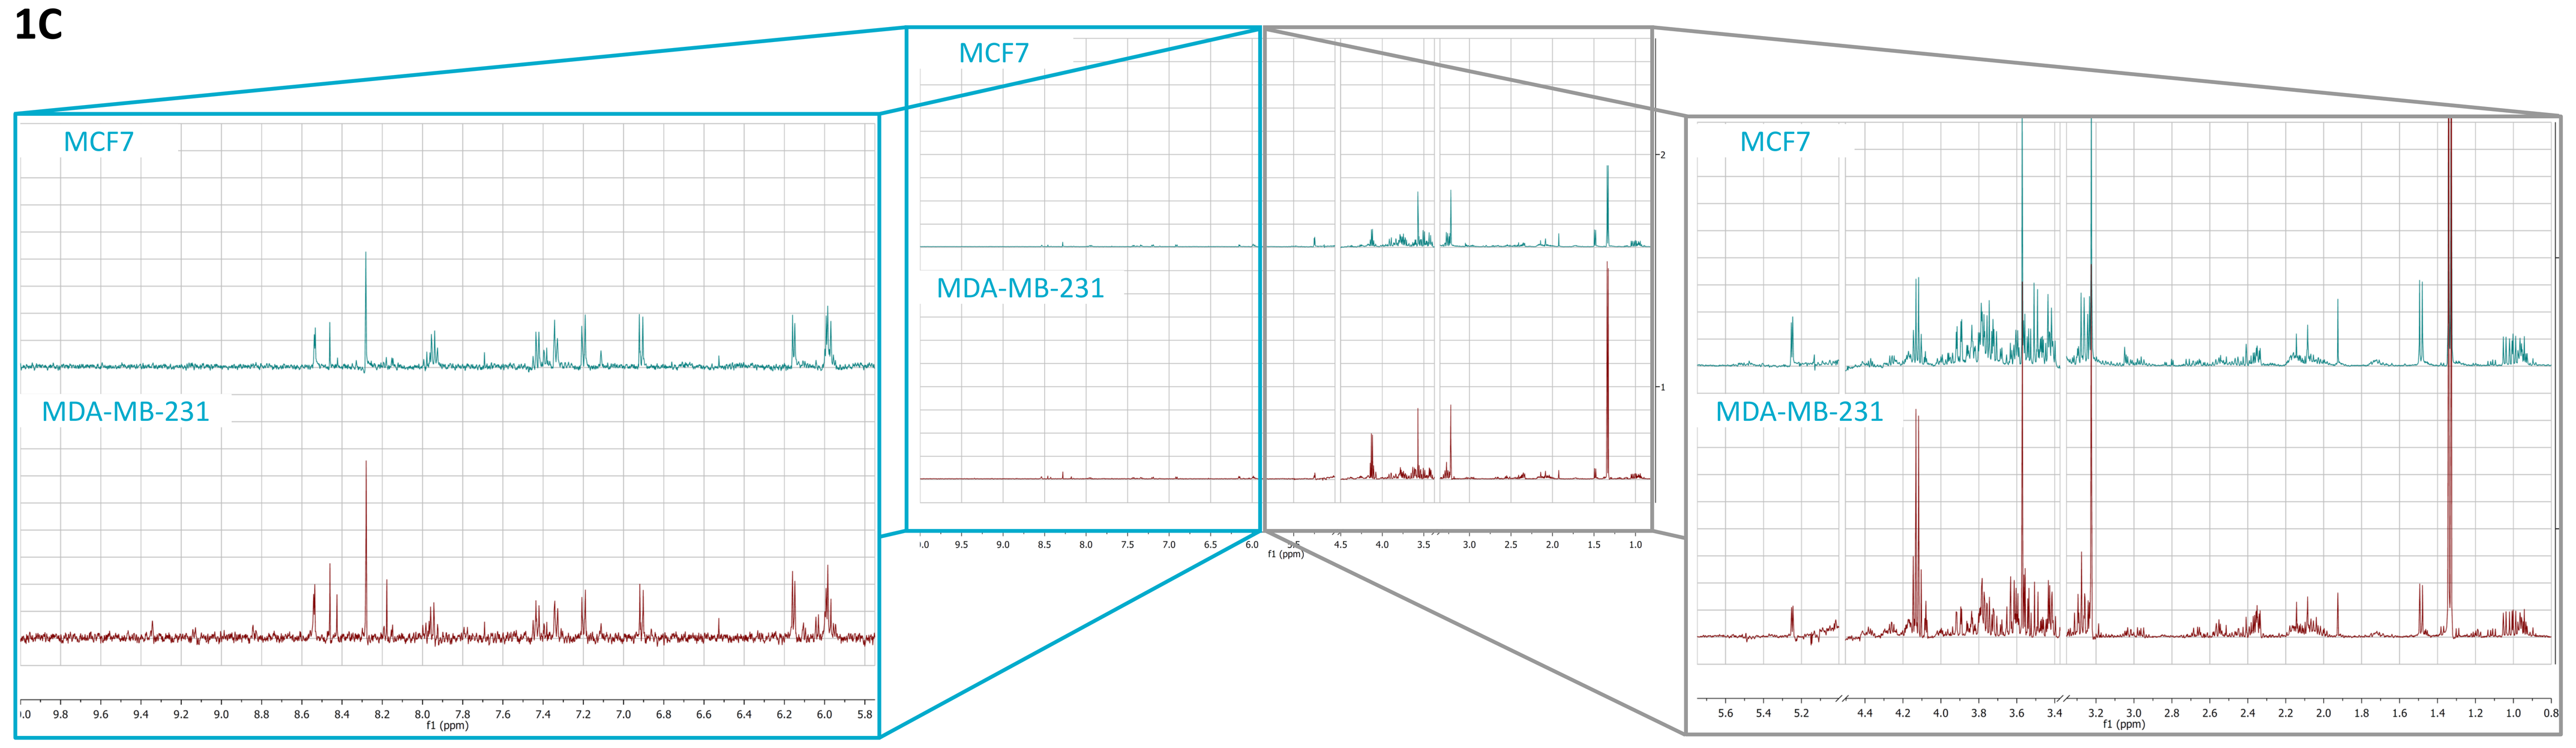
MCF7
MDA-MB-231
MCF7
MDA-MB-231
MCF7
MDA-MB-231
f1 (ppm)

1D

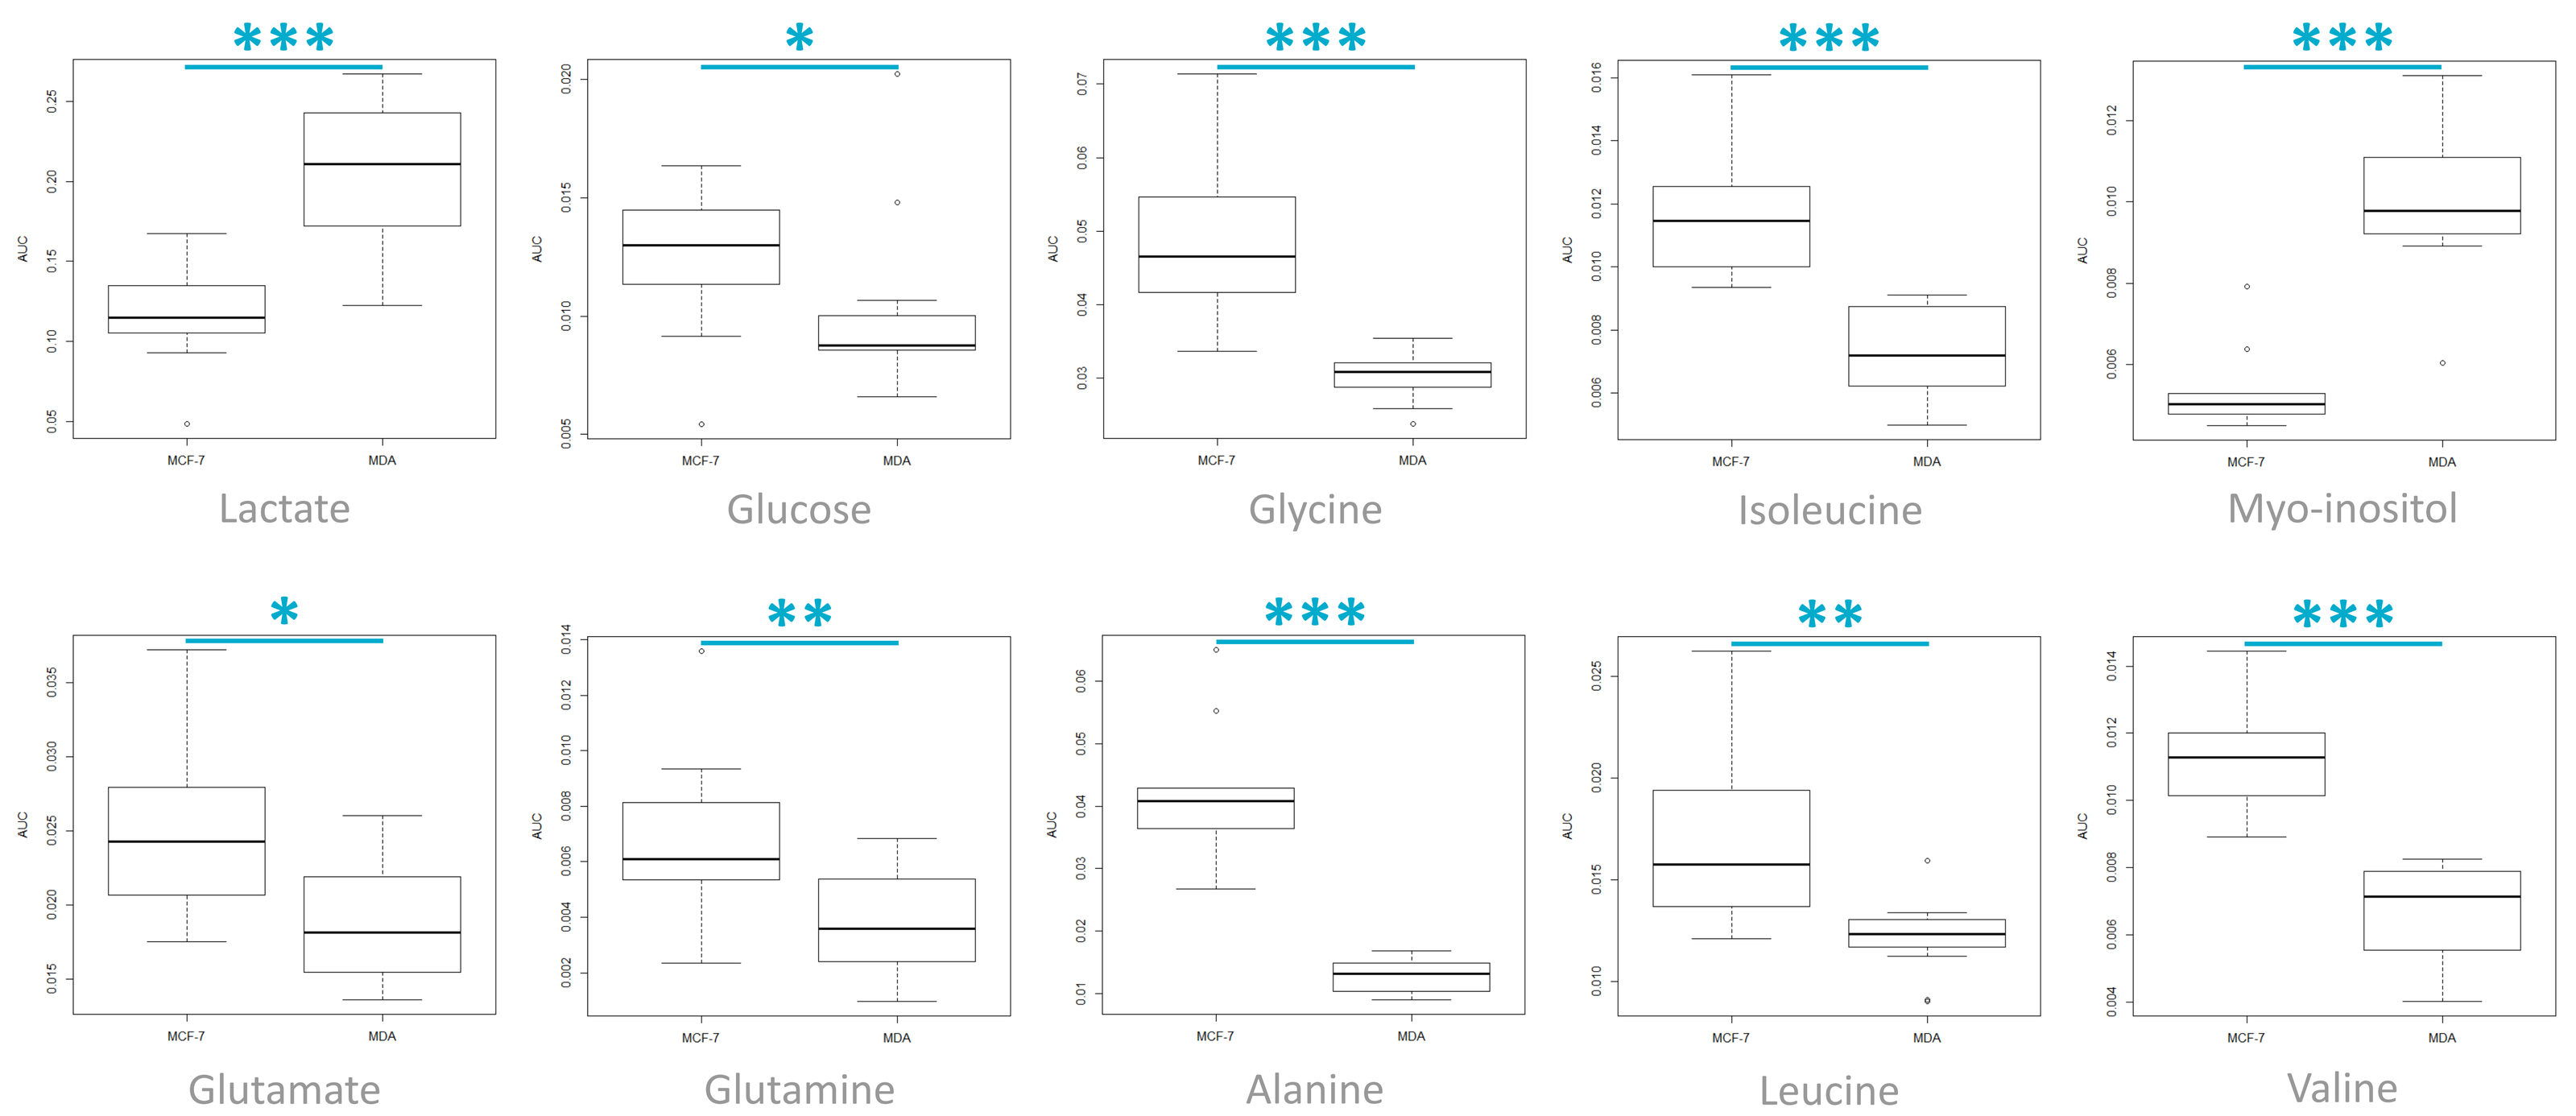

Lactate
*
Glucose

Glycine

Isoleucine

Myo-inositol
*
Glutamate
**
Glutamine

Alanine
**
Leucine

Valine

Fig-1A corresponds to score scatter plot obtained after PLS-DA on cell extracts (R^2X= 0.893; R^2Y= 0.932; Q^2= 0.871; hotelling T2: 0.95; CV-ANOVA: p=5.4x10^{-6}) note the difference between the composition of intracellular medium in MDA-MB-231 cells (black) and MCF7 cells (red).

The main discriminating metabolites between both cellular types are determined from the VIP value (Variable Importance in Projection) ≥1 (VIP plot; Fig-1B).

Fig-1C shows a comparison between ¹H-NMR spectra of cell extract for the MDA-MB-231 cells (bottom) and MCF7 cells (top). For better reading, the aromatic region (blue frame) and the aliphatic (gray frame) are zoomed.

Boxplots (Fig-1D) show a comparison of median Area Under the Curve (AUC) for metabolites with a VIP value ≥ 1 between MCF-7 and MDA-MB-231 cells.

(*: p-value<0.05; **: p-value<0.01; ***: p-value<0.001)

MCF7: MIF-sc vs MIF-KD

2A

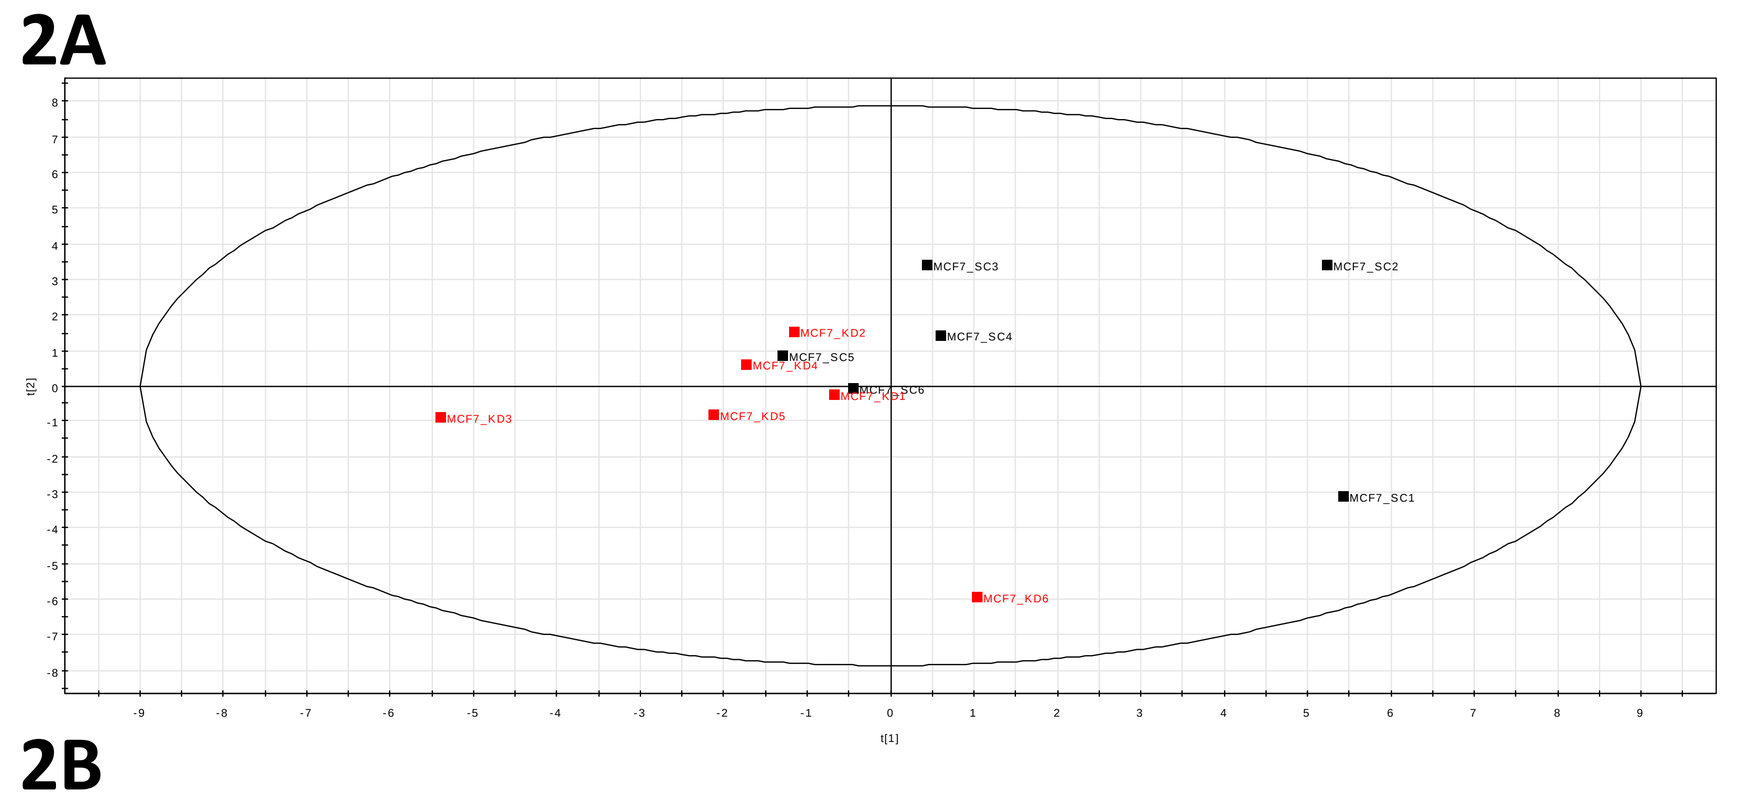

2C

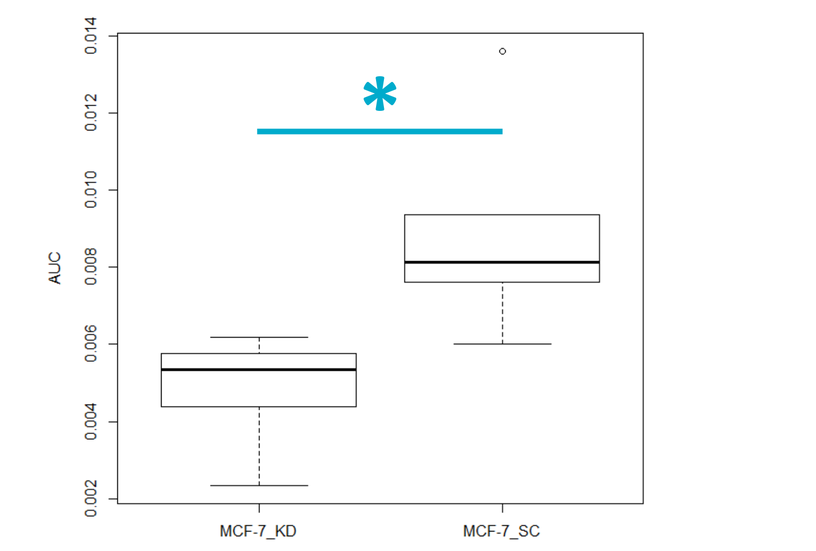
*
AUC
MCF-7_KD
MCF-7_SC
Glutamine

2B

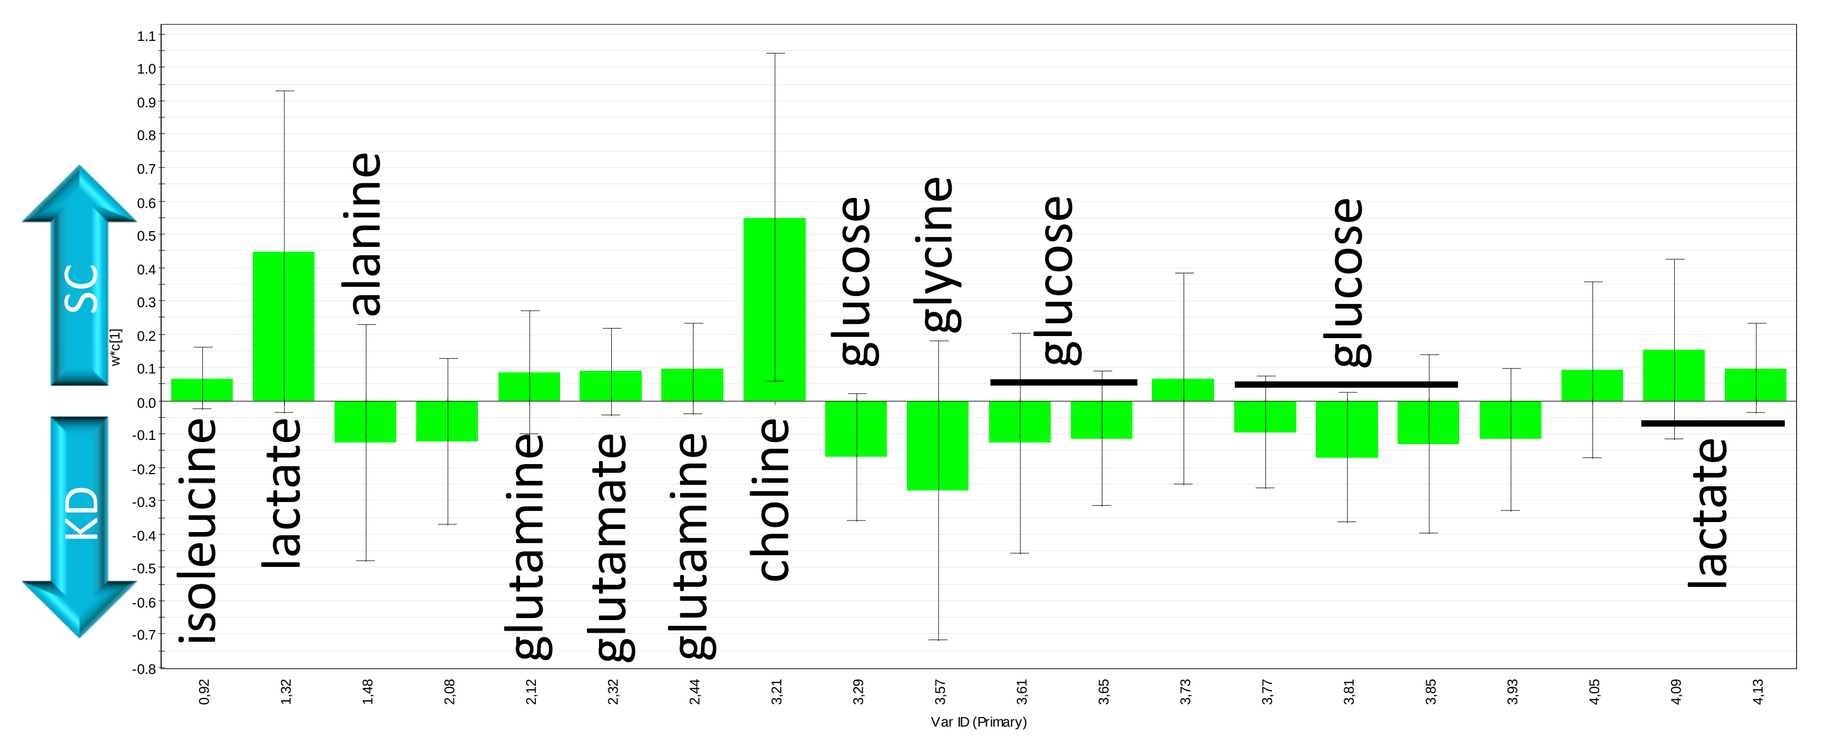
SC
KD
isoleucine
lactate
alanine
glutamine
glutamate
glutamine
choline
glucose
glycine
glucose
glucose
lactate
Var ID (Primary)

Fig-2A corresponds to score scatter plot obtained after PLS-DA on MCF7 cell extracts after MIF Knock down (red) or not (black) (R^2X= 0.79; R^2Y= 0.49; Q^2= -0.0936 ; hotelling T2 = 0.95). There is a non significant trend to separate the two condition (CV-ANOVA: p=1).

The loading column plot (Fig-2B) shows the discriminating metabolites (VIP value ≥1) between MIF-KD and MIF-sc in MCF-7 cell.

Boxplot (Fig-2C) show a comparison of median AUC for glutamine, the only significant discriminant metabolite (*: p-value<0.05).

MDA-MB-231: MIF-sc vs MIF-KD

3A

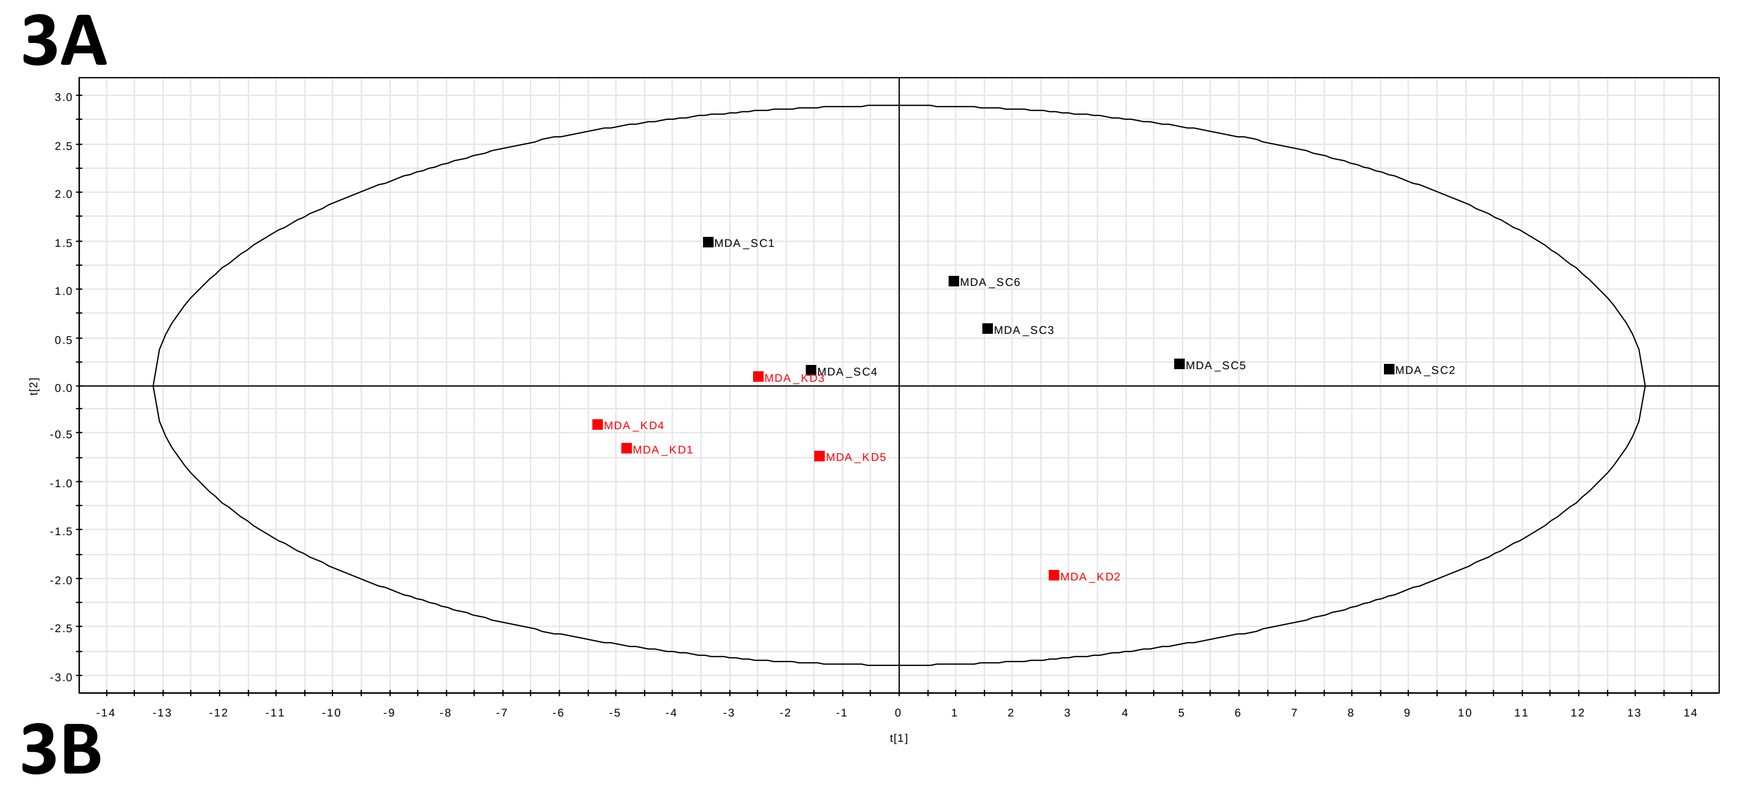

3C

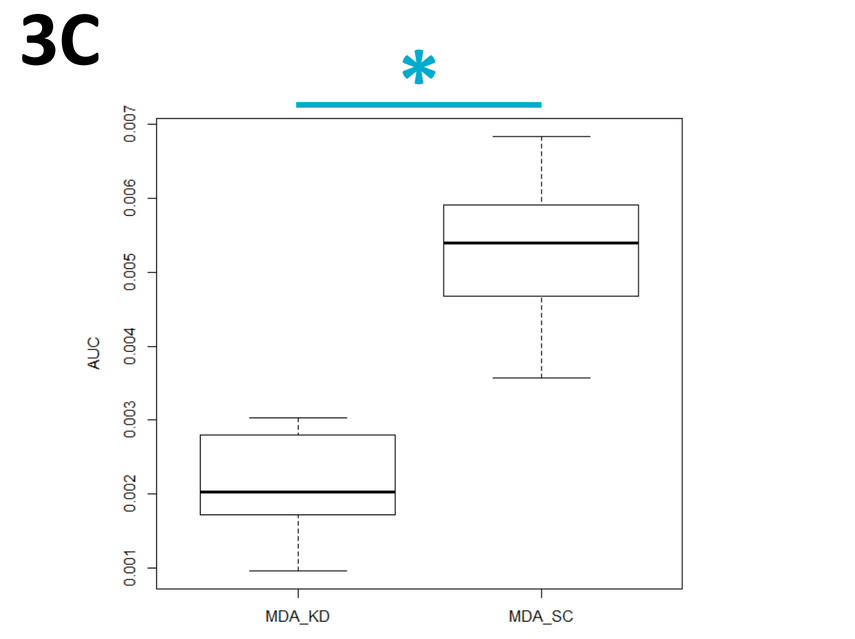
*
AUC
MDA_KD
MDA_SC
Glutamine

3B

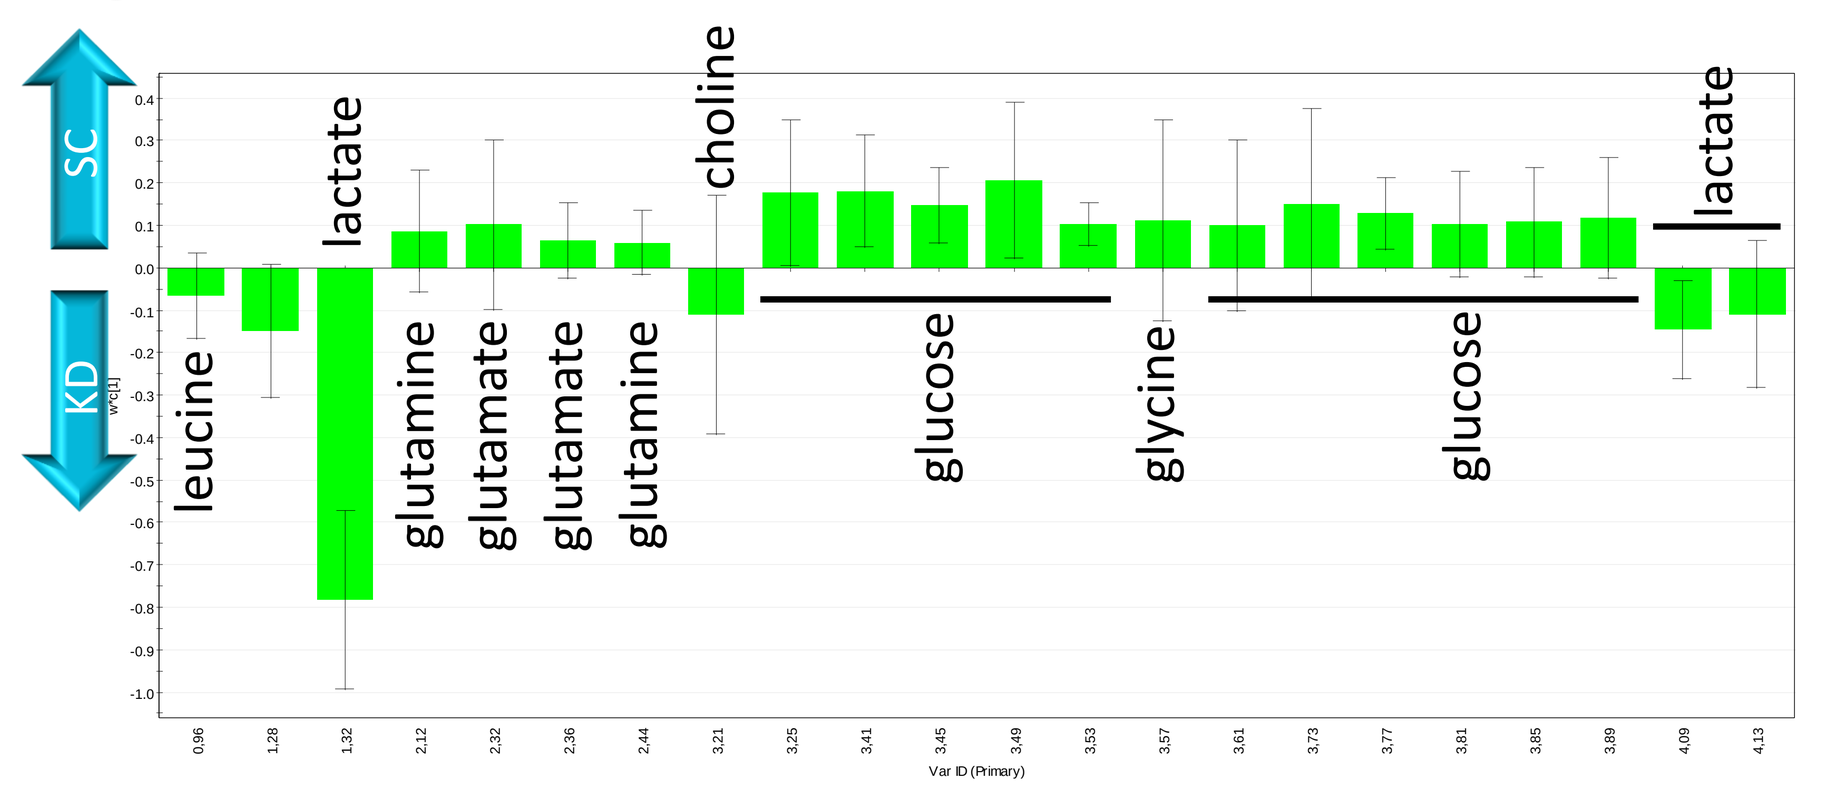
SC
KD
leucine
lactate
glutamine
glutamate
glutamate
glutamine
choline
glucose
glycine
glucose
lactate
Var ID (Primary)

Fig-3A corresponds to score scatter plot obtained after PLS-DA on MDA-MB-231 cell extracts after MIF Knock down (red) or not (black) (R^2X= 0.841; R^2Y= 0.815 ; Q^2= 0.192 ; hotelling T2 = 0.95). There is a non significant trend to separate the two condition (CV-ANOVA: p=0.93).

The loading column plot (Fig-3B) shows the discriminating metabolites (VIP value ≥1) between MIF-KD and MIF-sc in MDA-MB-231 cell.

Boxplot (Fig-3C) show a comparison of median AUC for glutamine, the only significant discriminant metabolite (*: p-value<0.05).

Conclusion

¹H-NMR spectroscopy of intracellular MDA-MB-231 and MCF-7 cells metabolomes shows significant different profiles suggesting that MDA-MB-231 cells have higher levels of glucose and glutamine consumptions than MCF-7, producing more lactate. This suggests that MDA-MB-231 cells are more dependent on glutamine but less on lactate and, possibly, different responses to targeted metabolic drugs. Moreover the decrease levels of glutamine observed in both cells lines when MIF is under-expressed suggests that this cytokine could be involved in the regulation of this amino acid metabolism.